Management of Lower Extremity Wounds in Clients High Risk for Wound Infection

Furqan Alex Khan, APRN ACNS-BC MSN, ALI A. PITAFI MD
www.ProHealthcare.us

Case 1

Trauma Wound | Left Shin | 3.2 x 4.5 x 0.2 cm
Patient Age: 88 | Gender: F
Healing Time: 6.5 Weeks

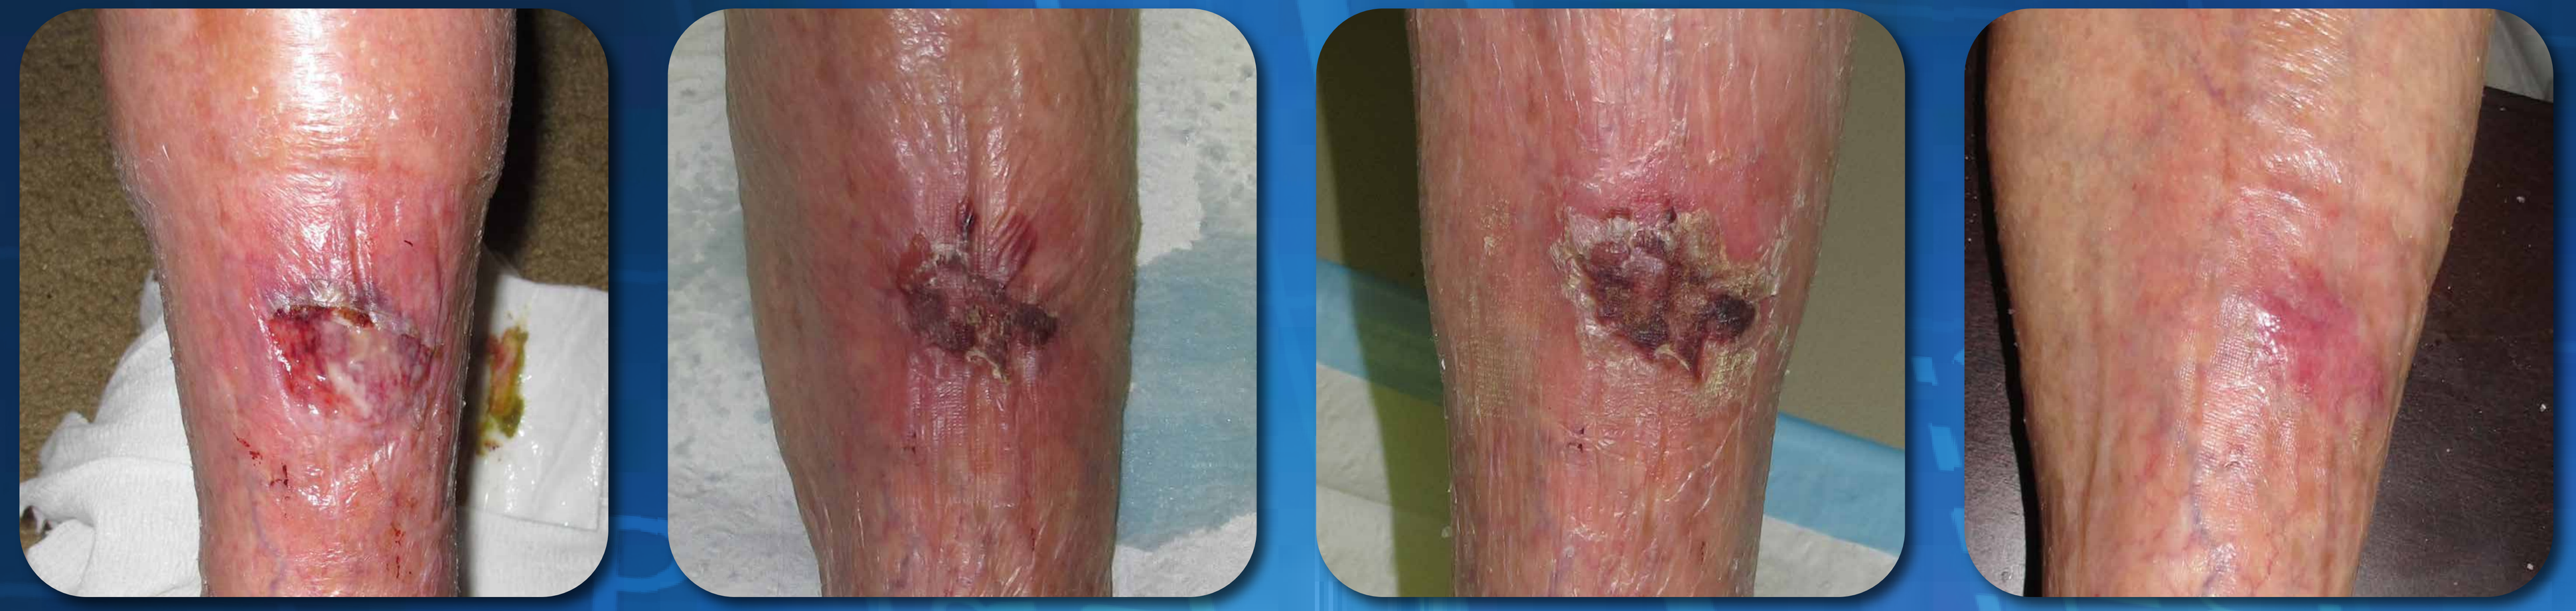

Case 2

Neuropathic Ulcer | Right Ankle | 2.1x 1.8 x 0.3 cm
Patient Age: 78 | Gender: M
Healing Time: 15.5 Weeks

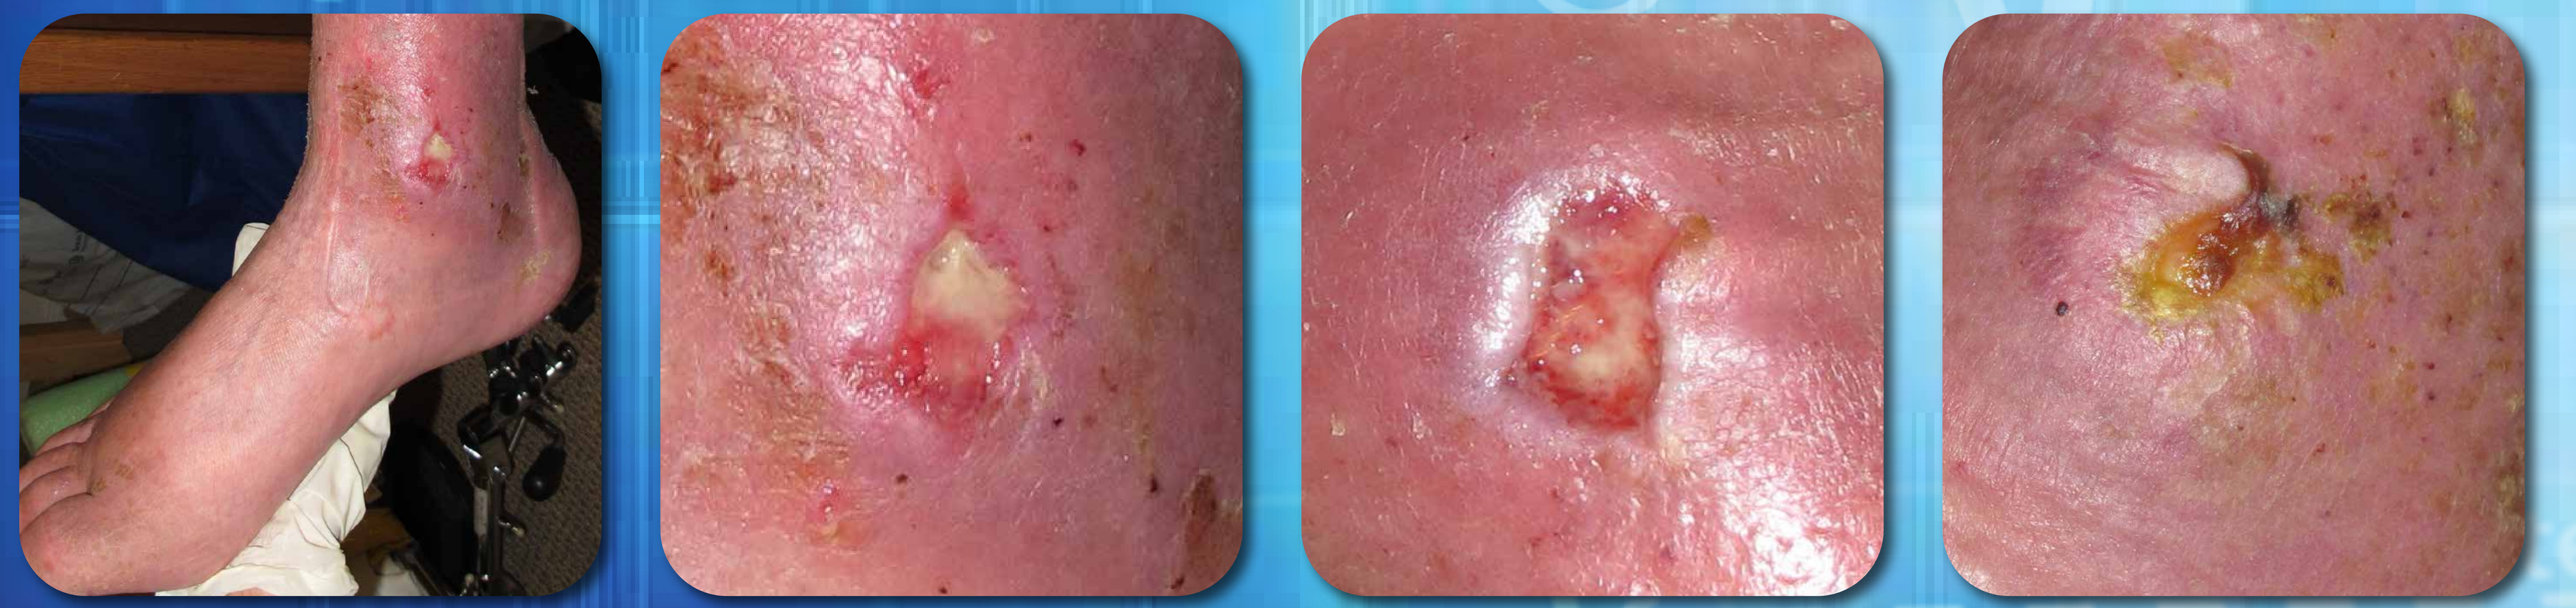

ANTI-MICROBIAL | ANTI-FUNGAL

ADVANCED WOUND CARE MANAGEMENT

PROBLEM

Infection of the lower extremity wounds is one of the most frequent problem identified in the homebound clients. In addition, wound infection not only alters the wound healing process but can lead to adverse outcomes, and mortality. Lower extremity ulcers are a major cause of morbidity in elderly clients and often colonized by many different microorganisms, including fungi. Some studies have identified the presence of Candida albicans, Candida parapsilosis, and Candida ciferrii in lower extremities wounds. Current clinical practice does not involve culturing of the wounds to identify presence of fungi; nor wounds are commonly treated with topical antifungal wound dressings. Further, wound cultures are commonly collected for aerobic and anaerobic bacterial identification and treated with oral antibiotics. Gentian violet is known to be antimycotic, antibacterial, and anthelmintic. It is potent against fungi like Candida, which causes various yeast infections, as well as bacteria including Streptococcus, Pseudomonas, and Staphylococcus, and even methicillin-resistant Staphylococcus aureus.

METHOD

Four (4) patients with the wounds of the lower extremity were identified who were receiving home health care for the management of their wounds. All four (4) patients have wounds with different etiology and were high risk for developing wound infection due to their complex comorbid medical conditions and their poor living conditions. All patients were treated with Polyurethane foam containing gentian violet, methylene blue, and silver. Wounds were cleansed with sterile water and 4x4 gauze each time and dressing was changed (3) times each week.

PRACTICE SETTING

All patients were treated in an outpatient clinic setting and examined by the treating practitioner once (1) a week.

RESULTS

Use of Polyurethane foam dressing with Methylene Blue, Gentian Violet, and Silver Sodium Zirconium Phosphate in all four (4) reviewed cases resulted in complete re-epithelialization of all wounds in less than 16 weeks. Patient made full recovery without any hospital admission. Sharp wound debridement was utilized to remove the non-viable tissue and Polyurethane foam dressing with Methylene Blue, Gentian Violet, and Silver Sodium Zirconium Phosphate dressing was utilized to create optimal wound healing environment by preventing wound infection, optimal moisture management and minimal pain with the dressing changes.

CONCLUSION

Use of Polyurethane foam with gentian violet, methylene blue, and silver was found to be effective in managing clients with lower extremities wounds.

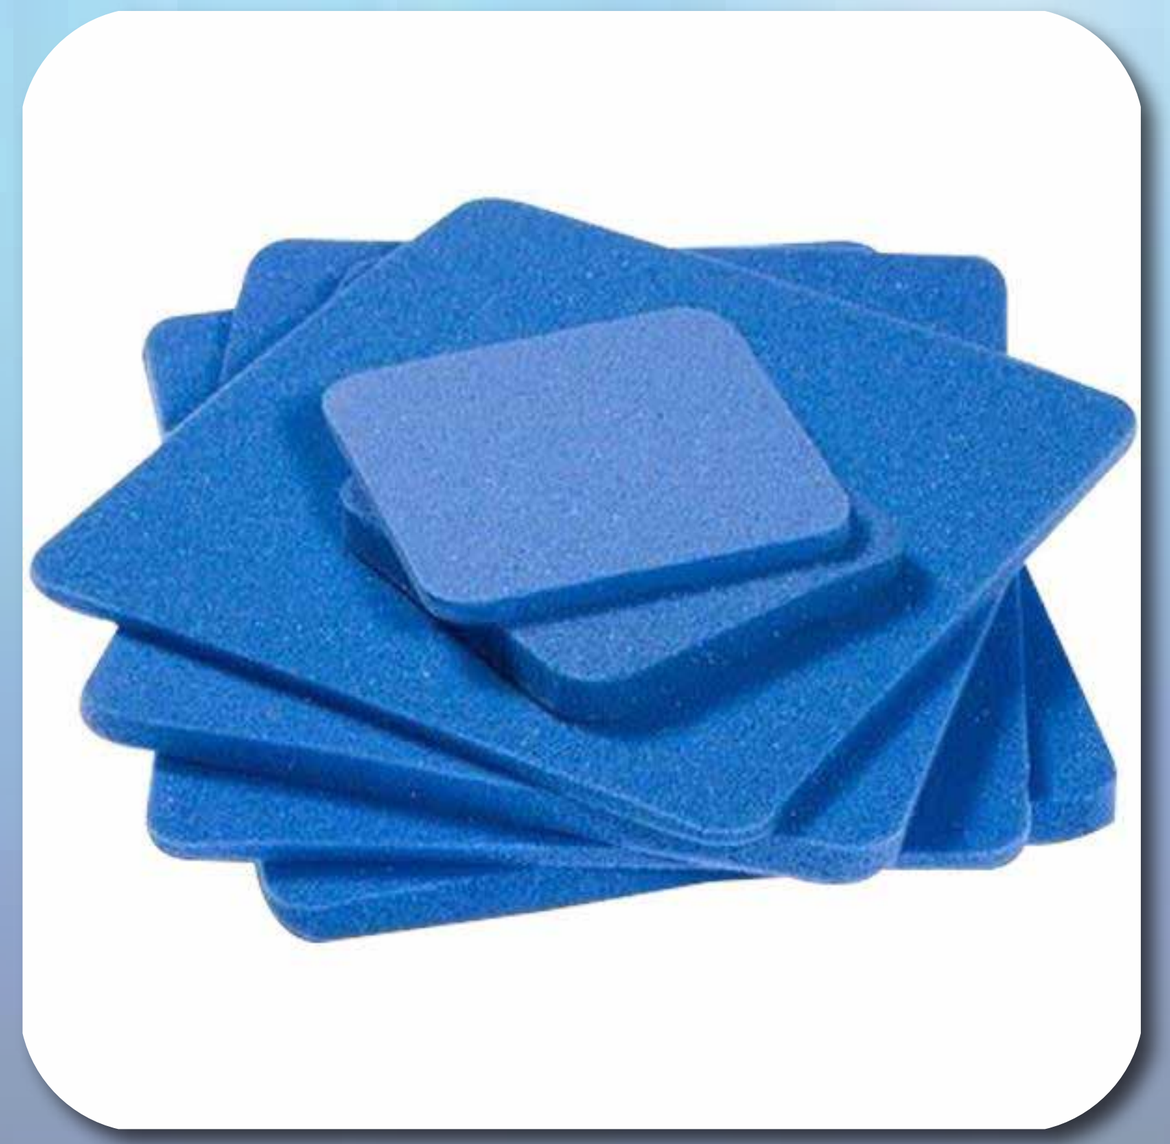

Case 3

Surgical Wound | Right Great Toe | 2.8 x 2.0 x 0.3 cm
Patient Age: 74 | Gender: M
Healing Time: 10.2 Weeks

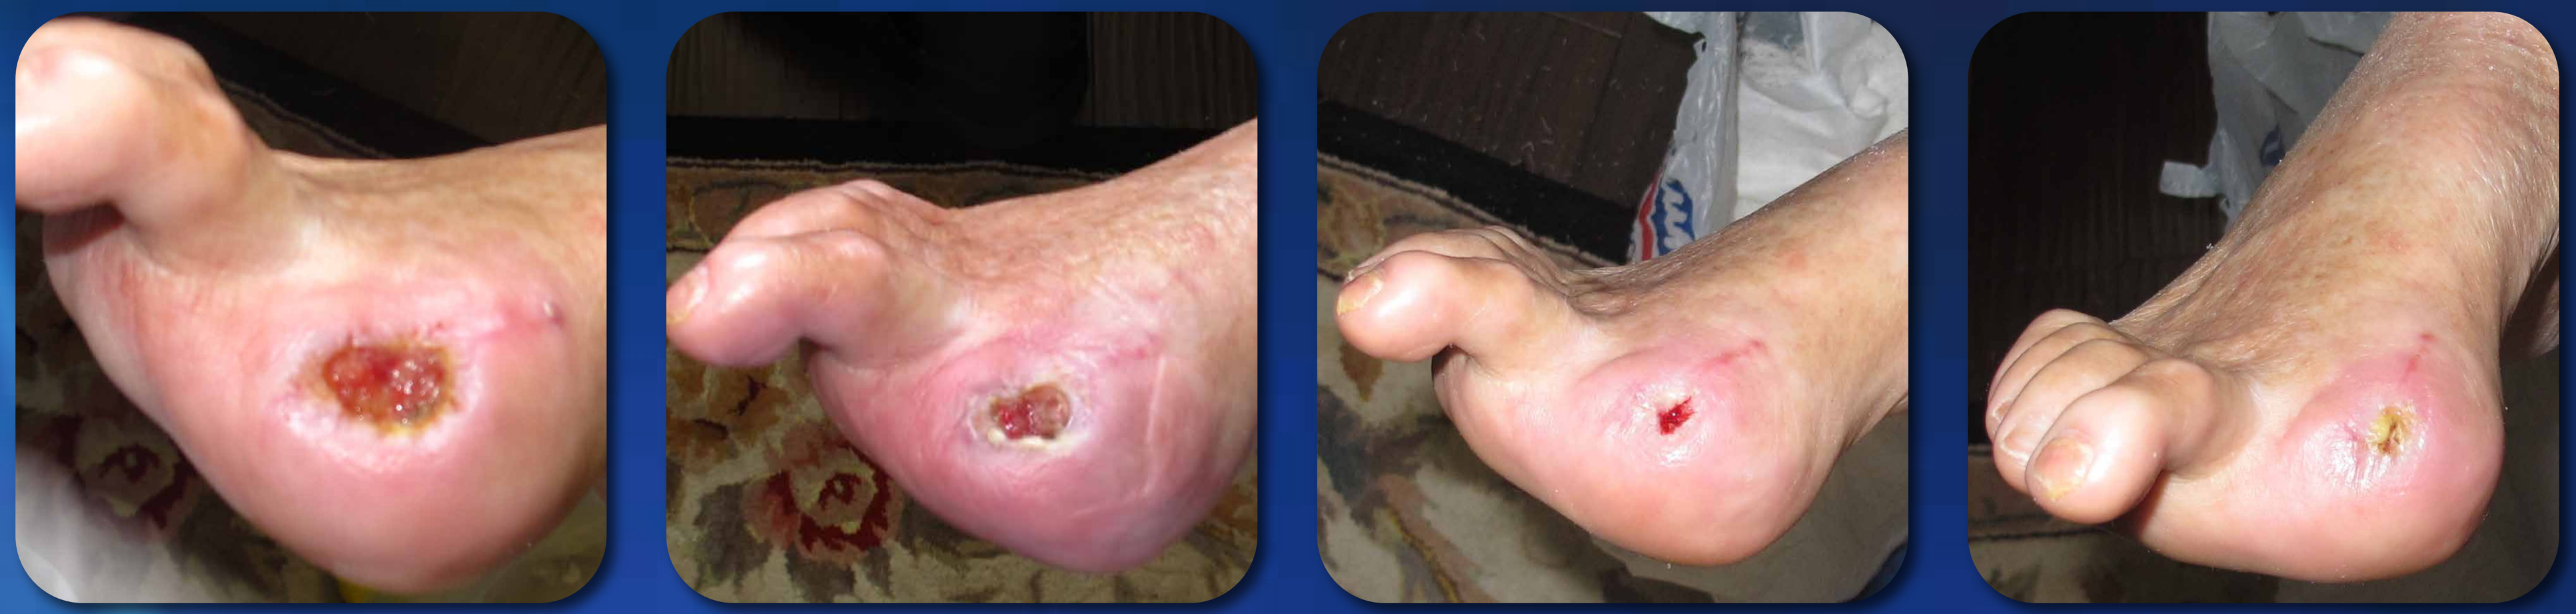

Case 4

Arterial Wound | Right Foot | Multiple Areas of Ulceration
Patient Age: 72 | Gender: F
Healing Time: 14 Weeks

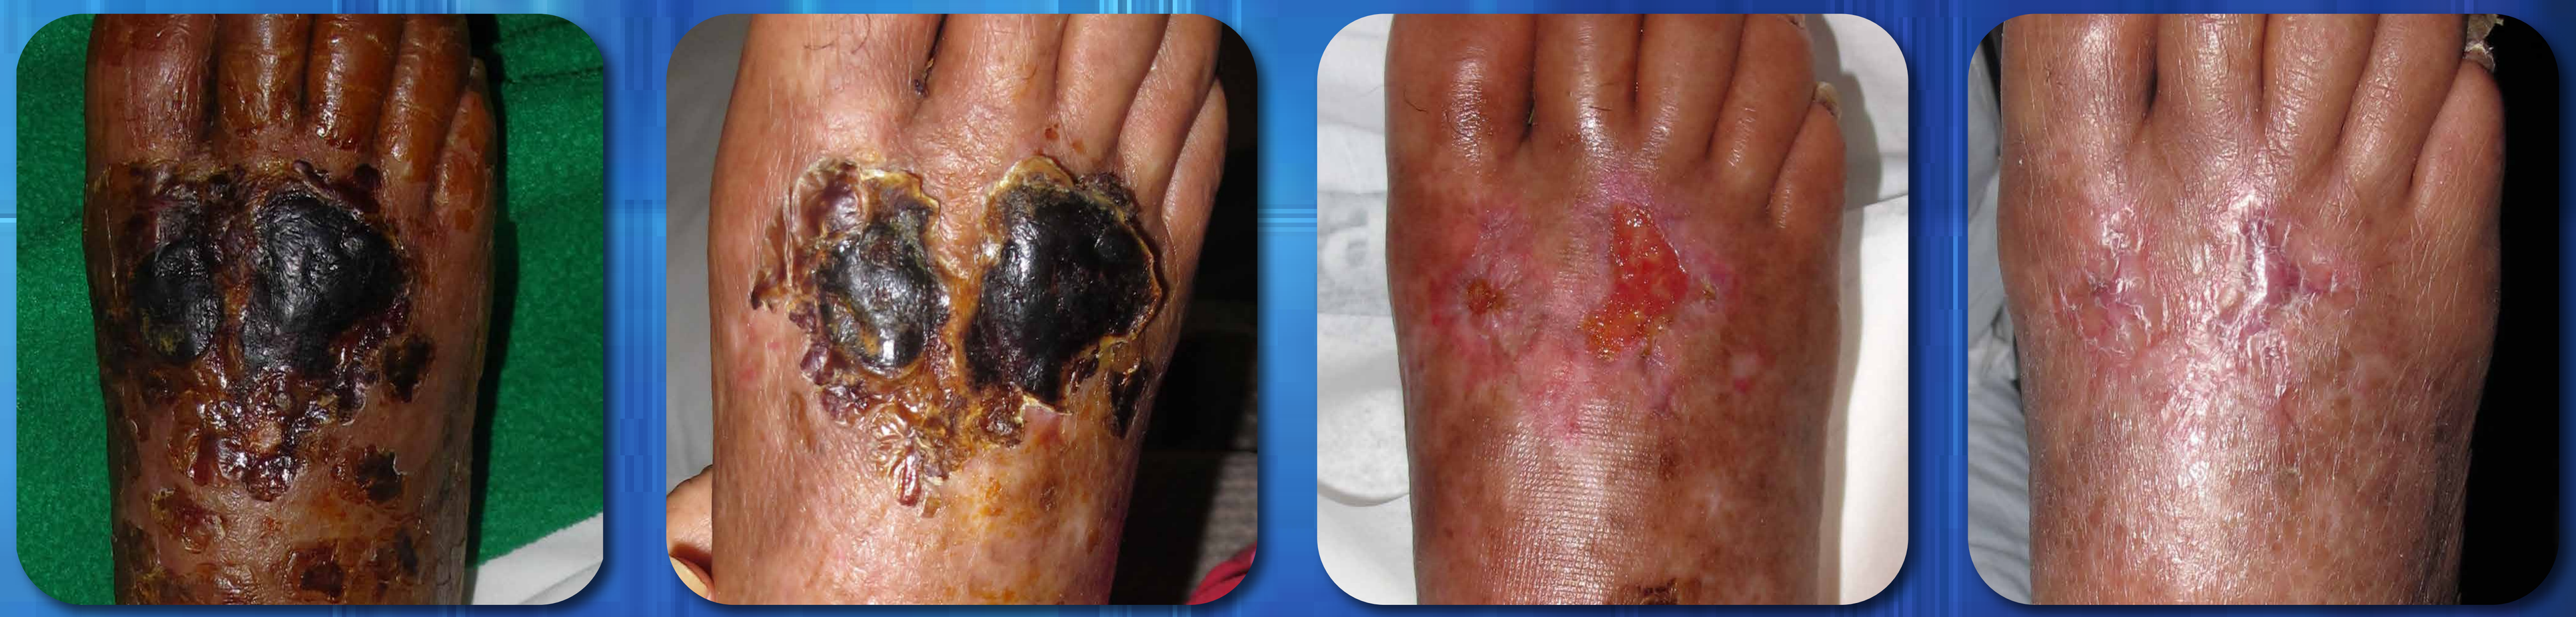

Bibliography:
S.Hsieh, E.Maranda, T.Saleh, M.Flores, J.Jimenez (2016). The Purple Dye That Heals. JAMA Dermatology April 2016.
C.Friedman, E.Bass, J.Steinberg (2006). Key Considerations For utilizing Silver Dressings. Podiatry Today May 2006.